EE366

carelonresearch.com

Real-World healthcare resource utilization and cost by relapse frequency among US patients with schizophrenia and schizoaffective disorder

Lia N. Pizzicato[1], Pin Xiang[2], Christopher L. Crowe[1], Chia-Chen Teng[1], Tristan Gloede[2], Yiling Yang[1], Joseph L. Smith[1], Keith Isenberg[3]

[1]Carelon Research, Inc., Wilmington, DE, [2]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, [3]Elevance Health, Inc., Indianapolis, IN

Background

- Schizophrenia and schizoaffective disorder are complex psychiatric conditions that are associated with substantial decrements in quality of life, healthcare resource utilization (HCRU), and costs.[1,2]
- Relapse, defined as acute exacerbations of symptoms, is common in schizophrenia and schizoaffective disorder.
- Gaps exist in our knowledge related to the extent of healthcare utilization and costs for patients experiencing relapses.[3] Moreover, it is unknown if HCRU and cost patterns differ between patients with commercial, Medicare Advantage, and Medicaid insurance.

Objectives

- To examine HCRU and costs among patients with schizophrenia and schizoaffective disorder by relapse frequency in three US payor populations (commercial, Medicare, and Medicaid).

Methods

- Patients ≥ 12 years with naïve schizophrenia or schizoaffective disorder (the earliest date is the index date) between 01/01/2014 and 08/31/2020 and continuous medical and pharmacy enrollment for one year before (baseline) and after index date (post-index) were identified from the Healthcare Integrated Research Database (HIRD®) **(Figure 1)** .

Figure 1. Patient selection

Step	Commercial N	Medicare N	Medicaid N	Description
1	30,855,158	30,855,158	4,886,682	Patients in the Healthcare Integrated Research Database (HIRD®) during the patient identification period (01/01/2014 and 08/31/2020).
2	45,161	45,161	30,068	From **Step 1**, patients with ≥ 1 inpatient/ER claim or ≥ 2 outpatient claims (≥ 30 days and ≤ 183 days apart) with a diagnosis of schizophrenia or schizoaffective disorder[1] during the patient identification period. Set the first diagnosis as the index date.
3	22,952	22,209	N/A	From **Step 2**, patients with Commercial or Medicare insurance on index date.
4	22,860	22,150	29,638	From **Step 3**, patients between 12 and 94 years on the index date.
5	21,351	16,844	27,291	From **Step 4**, patients without a neurocognitive disorder[2] during the study period (01/01/2013 to 08/31/2021).
6	17,412	14,022	22,983	From **Step 5**, those without a diagnosis for schizophrenia or schizoaffective disorder prior to the index date.
7	7,609	2,310	6,301	From **Step 6**, patients with ≥ 1 year of continuous medical and pharmacy enrollment prior to the index date.
8	4,974	1,660	4,858	From **Step 7**, patients with ≥ 1 year of continuous medical and pharmacy enrollment after the index date.
8a	3,726	1,178	3,048	Within **Step 8**, patients with 0 relapses during the post-index period.
8b	828	268	942	Within **Step 8**, patients with 1 relapse during the post-index period..
8c	420	214	868	Within **Step 8**, patients with ≥2 relapses during the post-index period..

1. Schizophrenia and schizoaffective disorder ICD-9-CM codes are 295.x and ICD-10-CM are F20.% and F25.%.
2. Neurocognitive disorders: Neurocognitive disorders ICD-9-CM codes are 290.4, 294.1, 294.11, 294.9, 331.83, 799.59, 331.x, 332.x and ICD-10-CM are F01.51, F01.50, F02.80, F02.81, F03.9%, G20.%, G30.%, G31.%, R41.9.

- Patients were stratified by relapse frequency within one-year post-index: 0 relapses, 1 relapse, and ≥2 relapses (defined as inpatient encounters for schizophrenia/schizoaffective disorder or emergency room encounters for psychiatric disorders).
- HCRU and costs were examined by relapse cohort and payor (commercial, Medicare Advantage/Supplemental, or Medicaid).
- Multivariable generalized linear models with gamma distribution and log-link functions were used to estimate adjusted post-index costs by relapse cohort.

Results

- Among 4,974 commercially insured patients, 75%, 17%, and 8% experienced 0, 1, and ≥2 relapses, respectively.
- Among 1,660 Medicare enrolled patients, 71%, 16%, and 13% experienced 0, 1, and ≥2 relapses, respectively.
- Among 4,858 Medicaid enrolled patients, 63%, 19%, and 18% experienced 0, 1, and ≥2 relapses, respectively.
- Commercial patients had less comorbidity than Medicare or Medicaid patients (**Table 1**).
- In general, patient characteristics were reflective of the populations served.

Table 1. Baseline patient demographic and clinical characteristics by number of relapses and payor

Demographic characteristics	Commercial			Medicare			Medicaid		
	0 Relapses	1 Relapse	≥2 Relapses	0 Relapses	1 Relapse	≥2 Relapses	0 Relapses	1 Relapse	≥2 Relapses
Number of patients, N	3,726	828	420	1,178	268	214	3,048	942	868
Demographics									
Age on index (years), mean (SD)	33.5 (14.67)	30.3 (13.04)	29.4 (12.45)	56.6 (13.47)	52.3 (11.92)	49.9 (11.74)	34.9 (14.75)	34.5 (13.62)	34.8 (12.11)
Female, n (%)	1,644 (44.1%)	328 (39.6%)	199 (47.4%)	583 (49.5%)	129 (48.1%)	113 (52.8%)	1,561 (51.2%)	521 (55.3%)	446 (51.4%)
Geographic region of patient,[1] n (%)									
Northeast	490 (13.2%)	136 (16.4%)	63 (15.0%)	239 (20.3%)	55 (20.5%)	35 (16.4%)	0 (0%)	0 (0%)	0 (0%)
Midwest	874 (23.5%)	179 (21.6%)	82 (19.5%)	511 (43.4%)	140 (52.2%)	99 (46.3%)	0 (0%)	0 (0%)	0 (0%)
South	1,196 (32.1%)	268 (32.4%)	128 (30.5%)	222 (18.8%)	53 (19.8%)	45 (21.0%)	2,334 (76.6%)	735 (78.0%)	636 (73.3%)
West	1,058 (28.4%)	218 (26.3%)	132 (31.4%)	206 (17.5%)	20 (7.5%)	35 (16.4%)	714 (23.4%)	207 (22.0%)	232 (26.7%)
Rural/urban designation,[1] n (%)									
Rural	972 (26.1%)	213 (25.7%)	106 (25.2%)	215 (18.3%)	47 (17.5%)	38 (17.8%)	511 (16.8%)	156 (16.6%)	140 (16.1%)
Urban	2,719 (73.0%)	606 (73.2%)	312 (74.3%)	962 (81.7%)	221 (82.5%)	176 (82.2%)	2,537 (83.2%)	786 (83.4%)	728 (83.9%)
Year of index date, n (%)									
2014	590 (15.8%)	158 (19.1%)	78 (18.6%)	146 (12.4%)	37 (13.8%)	29 (13.6%)	202 (6.6%)	69 (7.3%)	87 (10.0%)
2015	602 (16.2%)	129 (15.6%)	78 (18.6%)	123 (10.4%)	33 (12.3%)	11 (5.1%)	330 (10.8%)	108 (11.5%)	102 (11.8%)
2016	630 (16.9%)	143 (17.3%)	62 (14.8%)	147 (12.5%)	41 (15.3%)	30 (14.0%)	422 (13.8%)	122 (13.0%)	86 (9.9%)
2017	558 (15.0%)	134 (16.2%)	61 (14.5%)	168 (14.3%)	39 (14.6%)	37 (17.3%)	343 (11.3%)	125 (13.3%)	108 (12.4%)
2018	508 (13.6%)	97 (11.7%)	59 (14.0%)	180 (15.3%)	25 (9.3%)	34 (15.9%)	437 (14.3%)	173 (18.4%)	140 (16.1%)
2019	535 (14.4%)	101 (12.2%)	51 (12.1%)	263 (22.3%)	57 (21.3%)	52 (24.3%)	728 (23.9%)	191 (20.3%)	210 (24.2%)
2020[2]	303 (8.1%)	66 (8.0%)	31 (7.4%)	151 (12.8%)	36 (13.4%)	21 (9.8%)	586 (19.2%)	154 (16.3%)	135 (15.6%)
Social determinants of health (SDoH) – census block level									
Median family income in past 12 months, mean (SD)	$86,898 ($44,947)	$87,926 ($45,557)	$89,040 ($45,164)	$75,380 ($39,589)	$70,335 ($43,652)	$71,782 ($40,760)	$63,903 ($33,134)	$58,922 ($31,090)	$58,979 ($31,247)
Proportion of population who are non-Hispanic black, mean (SD)	0.11 (0.20)	0.11 (0.20)	0.10 (0.17)	0.17 (0.26)	0.20 (0.28)	0.18 (0.27)	0.36 (0.32)	0.39 (0.33)	0.36 (0.32)
Proportion of population who are Hispanic/Latino, mean (SD)	0.12 (0.18)	0.12 (0.18)	0.13 (0.19)	0.13 (0.20)	0.11 (0.19)	0.11 (0.18)	0.13 (0.17)	0.13 (0.18)	0.14 (0.19)
Quartiles of socioeconomic status (SES),[1,3] n (%)									
1 (Low SES)	615 (16.5%)	144 (17.4%)	64 (15.2%)	296 (25.1%)	86 (32.1%)	76 (35.5%)	1,265 (41.5%)	435 (46.2%)	398 (45.9%)
2	767 (20.6%)	158 (19.1%)	74 (17.6%)	292 (24.8%)	72 (26.9%)	48 (22.4%)	771 (25.3%)	229 (24.3%)	207 (23.8%)
3	938 (25.2%)	211 (25.5%)	98 (23.3%)	299 (25.4%)	55 (20.5%)	45 (21.0%)	484 (15.9%)	128 (13.6%)	135 (15.6%)
4 (High SES)	1,323 (35.5%)	299 (36.1%)	172 (41.0%)	256 (21.7%)	47 (17.5%)	43 (20.1%)	333 (10.9%)	60 (6.4%)	54 (6.2%)
Clinical characteristic and comorbidities									
Quan-Charlson comorbidity index (QCI), mean (SD)	0.3 (0.88)	0.3 (0.78)	0.5 (1.07)	1.0 (1.59)	1.0 (1.44)	1.2 (1.67)	0.5 (1.06)	0.6 (1.13)	0.8 (1.52)
Asthma	298 (8.0%)	84 (10.1%)	65 (15.5%)	119 (10.1%)	53 (19.8%)	52 (24.3%)	407 (13.4%)	158 (16.8%)	173 (19.9%)
Dyslipidemia	580 (15.6%)	118 (14.3%)	59 (14.0%)	531 (45.1%)	113 (42.2%)	104 (48.6%)	512 (16.8%)	168 (17.8%)	150 (17.3%)
Hypertension	612 (16.4%)	139 (16.8%)	79 (18.8%)	595 (50.5%)	144 (53.7%)	108 (50.5%)	805 (26.4%)	278 (29.5%)	310 (35.7%)
Overweight/obesity	497 (13.3%)	94 (11.4%)	75 (17.9%)	294 (25.0%)	77 (28.7%)	67 (31.3%)	576 (18.9%)	197 (20.9%)	205 (23.6%)
Behavioral health comorbidities, n (%)									
Alcohol use disorder	299 (8.0%)	121 (14.6%)	68 (16.2%)	71 (6.0%)	30 (11.2%)	37 (17.3%)	260 (8.5%)	124 (13.2%)	195 (22.5%)
Anxiety disorders	1,523 (40.9%)	361 (43.6%)	232 (55.2%)	328 (27.8%)	101 (37.7%)	100 (46.7%)	1,000 (32.8%)	386 (41.0%)	453 (52.2%)
Attention-deficit/hyperactivity disorder	478 (12.8%)	134 (16.2%)	82 (19.5%)	39 (3.3%)	≤10	≤10	448 (14.7%)	130 (13.8%)	117 (13.5%)
Bipolar disorder	1,020 (27.4%)	274 (33.1%)	187 (44.5%)	314 (26.7%)	102 (38.1%)	107 (50.0%)	883 (29.0%)	383 (40.7%)	417 (48.0%)
Depression	1,532 (41.1%)	373 (45.0%)	237 (56.4%)	399 (33.9%)	101 (37.7%)	103 (48.1%)	1,231 (40.4%)	437 (46.4%)	470 (54.1%)
Emotional disturbance	67 (1.8%)	18 (2.2%)	21 (0.5%)	≤10	≤10	≤10	55 (1.8%)	21 (2.2%)	26 (3.0%)
Intellectual disability	37 (1.0%)	≤10	≤10	29 (2.5%)	≤10	≤10	96 (3.1%)	22 (2.3%)	29 (3.3%)
Learning disability	18 (0.5%)	≤10	≤10	≤10	≤10	0 (0.0%)	30 (1.0%)	≤10	≤10
Nicotine dependence	552 (14.8%)	211 (25.5%)	144 (34.3%)	269 (22.8%)	101 (37.7%)	101 (47.2%)	911 (29.9%)	364 (38.6%)	511 (58.9%)
Personality disorder	173 (4.6%)	58 (7.0%)	55 (13.1%)	36 (3.1%)	20 (7.5%)	27 (12.6%)	120 (3.9%)	65 (6.9%)	75 (8.6%)
Post traumatic stress disorder	71 (1.9%)	22 (2.7%)	32 (7.6%)	14 (1.2%)	≤10	≤10	48 (1.6%)	17 (1.8%)	32 (3.7%)
Substance use disorder	664 (17.8%)	227 (27.4%)	140 (33.3%)	129 (11.0%)	44 (16.4%)	62 (29.0%)	623 (20.4%)	318 (33.8%)	417 (48.0%)
Suicidal attempts/injuries related to intentional self-harm	75 (2.0%)	20 (2.4%)	25 (6.0%)	14 (1.2%)	≤10	≤10	47 (1.5%)	41 (4.4%)	46 (5.3%)
Suicidal ideation	456 (12.2%)	157 (19.0%)	128 (30.5%)	60 (5.1%)	35 (13.1%)	45 (21.0%)	391 (12.8%)	211 (22.4%)	264 (30.4%)

[1]The categories may not sum to 100% due to missing values.
[2]The socioeconomic status (SES) index is a composite measure based on seven SDoH variables. A score of 4 indicates the patient is in the top 25% of SES and a score of 1 indicates the patient is in the bottom 25% of SES using all Census block groups and 2017 as the reference basis for calculation. Patients unable to be linked to SDOH data or who have at least one of the seven variables missing are categorized as unknown.
[3]Data was only available through August 31, 2020.

- For all payors, the proportion of patients with inpatient, emergency room, and office visits increased with an increasing number of relapses (**Figures 2-4**).
- For all payors, the proportion of patients with visits increased across settings between baseline and post-index, except for emergency room visits.

Figure 2. Proportion of patients with ≥1 inpatient visit during baseline and post-index by relapse and payor

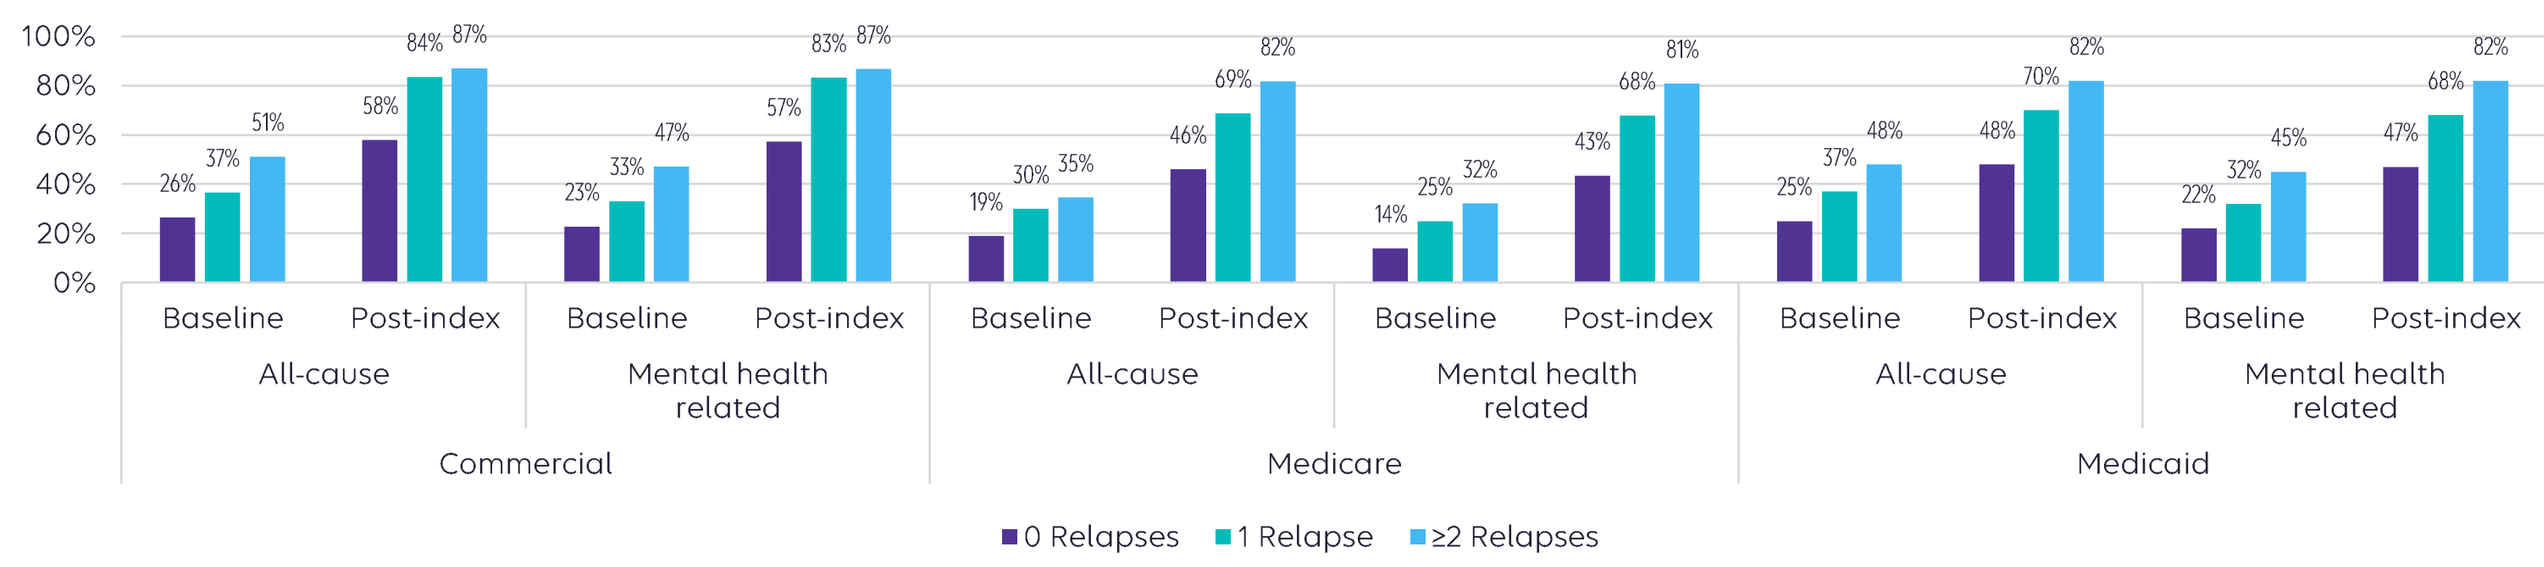
100%
80%
60%
40%
20%
0%
26% 37% 51%
58% 84% 87%
23% 33% 47%
57% 83% 87%
19% 30% 35%
46% 69% 82%
14% 25% 32%
43% 68% 81%
25% 37% 48%
48% 70% 82%
22% 32% 45%
47% 68% 82%
Baseline
Post-index
All-cause
Mental health related
Commercial
Medicare
Medicaid
0 Relapses
1 Relapse
≥2 Relapses

Figure 3. Proportion of patients with ≥1 emergency room visit during baseline and post-index by relapse and payor

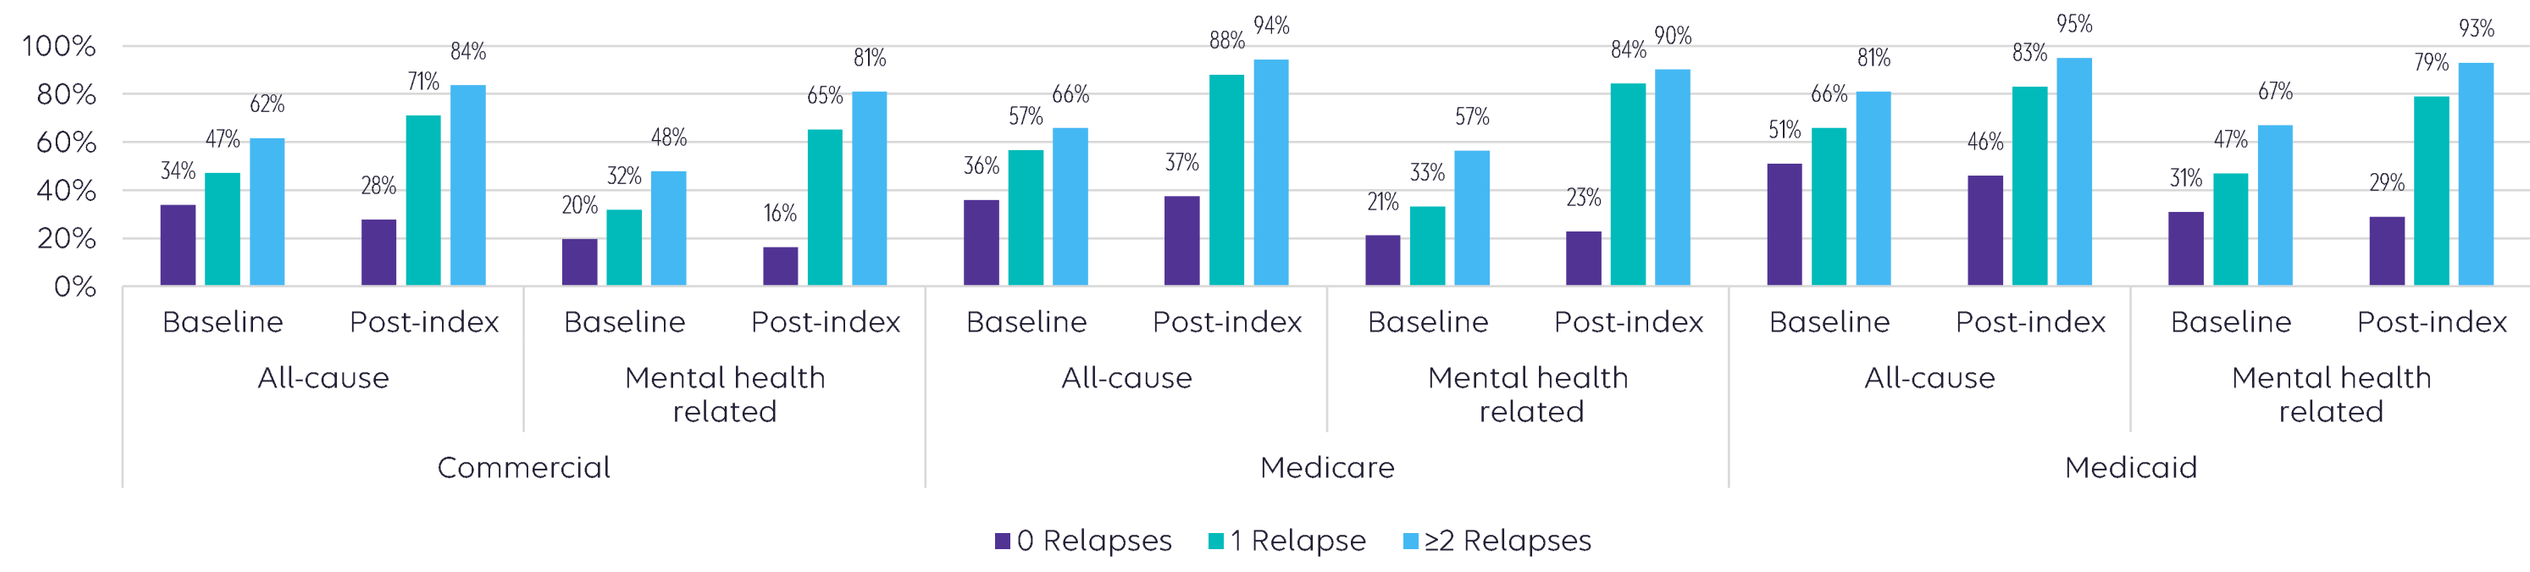
100%
80%
60%
40%
20%
0%
34% 47% 62%
28% 71% 84%
20% 32% 48%
16% 65% 81%
36% 57% 66%
37% 88% 94%
21% 33% 57%
23% 84% 90%
51% 66% 81%
46% 83% 95%
31% 47% 67%
29% 79% 93%
Baseline
Post-index
All-cause
Mental health related
Commercial
Medicare
Medicaid
0 Relapses
1 Relapse
≥2 Relapses

Figure 4. Proportion of patients with ≥1 office visit during baseline and post-index by number of relapses and payor

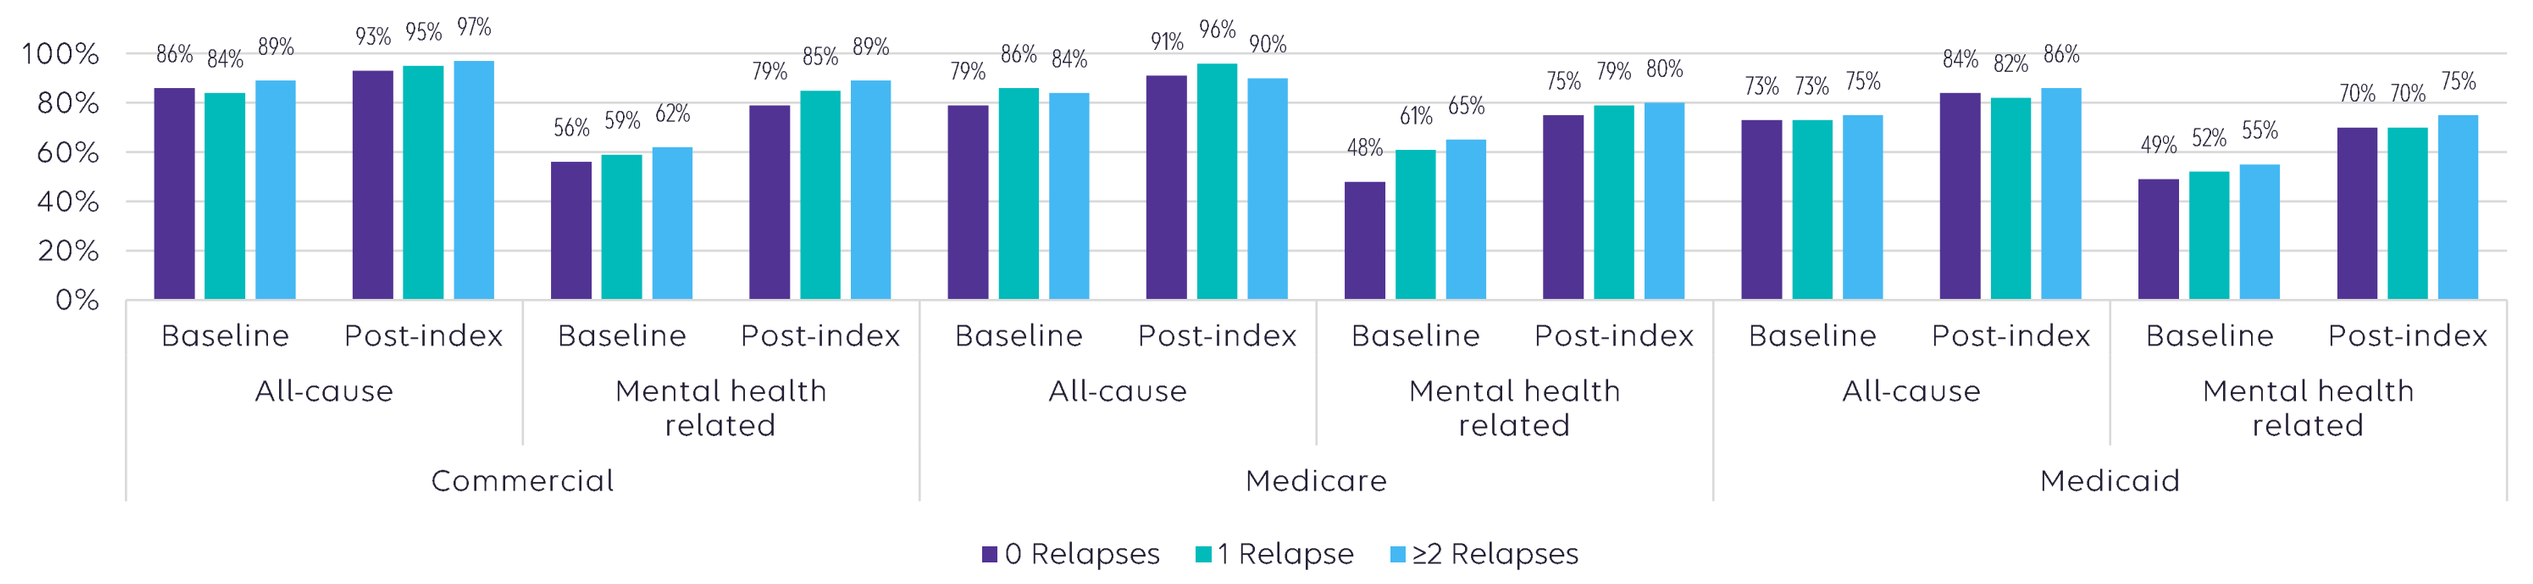
100%
80%
60%
40%
20%
0%
86% 84% 89%
93% 95% 97%
56% 59% 62%
79% 85% 89%
79% 86% 84%
91% 96% 90%
48% 61% 65%
75% 79% 80%
73% 73% 75%
84% 82% 86%
49% 52% 55%
70% 70% 75%
Baseline
Post-index
All-cause
Mental health related
Commercial
Medicare
Medicaid
0 Relapses
1 Relapse
≥2 Relapses

- Two-thirds of commercial, one-third of Medicare, and less than half of Medicaid patients had at least one psychotherapy visit on or after the index date (**Figure 5**).
 - Of patients who had ≥1 psychotherapy visit, 50% of commercial received 9 or fewer visits and 50% of Medicare and Medicaid patients received 5 or fewer visits.
 - The number of visits tended to decrease with more relapses.

Figure 5. Post-index psychotherapy utilization by number of relapses and payor

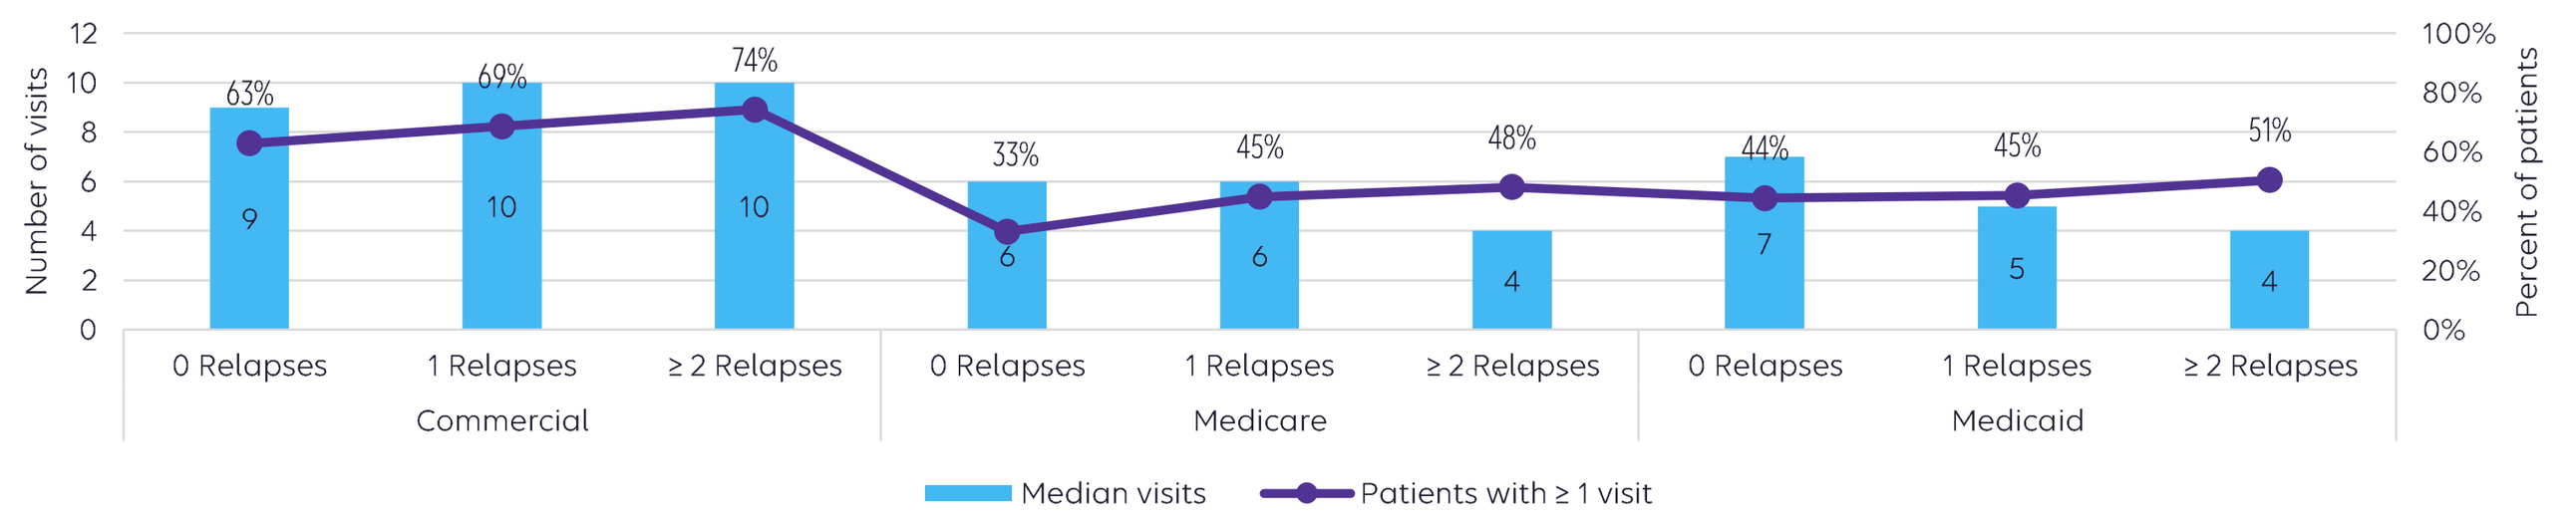
Number of visits
Percent of patients
63% 69% 74% 33% 45% 48% 44% 45% 51%
9 10 10 6 6 4 7 5 4
0 Relapses
1 Relapses
≥ 2 Relapses
Commercial
Medicare
Medicaid
Median visits
Patients with ≥ 1 visit

- Unadjusted total cost increased from pre-index to post-index by between 55% and 158% across all relapse cohorts and payors (**Table 2**).

Table 2. All-cause and mental health related unadjusted total costs (medical+pharmacy in 2020$) by relapse and payor

	Commercial			Medicare			Medicaid		
	0 Relapses	1 Relapse	≥2 Relapses	0 Relapses	1 Relapse	≥2 Relapses	0 Relapses	1 Relapse	≥2 Relapses
Number of patients, N	3,726	828	420	1,178	268	214	3,048	942	868
Total costs									
Baseline	$18,608	$25,119	$36,284	$16,053	$20,705	$25,809	$10,726	$14,711	$19,099
Post-index	$33,382	$63,151	$93,483	$24,825	$38,612	$47,472	$17,122	$25,779	$40,358
Mental health related									
Baseline	$11,403	$18,252	$26,473	$6,813	$10,364	$15,991	$6,185	$8,492	$12,709
Post-index	$26,196	$56,680	$81,900	$14,215	$26,901	$36,563	$12,387	$19,209	$33,319

- In adjusted models, post-index mean total costs were significantly greater among those with 1 and ≥2 relapses than 0 relapses for all payors, for all-cause and mental health related costs (**Figure 7**).
- Adjusted all–cause total costs increased with increasing number of relapses and were highest in commercial patients ($32,786; $57,501; and $79,651 for 0, 1, and ≥2 relapses, respectively) relative to Medicare ($29,301; $44,292; and $48,185) and Medicaid ($19,416; $28,219; and $41,929).

Figure 7. Adjusted mental health related total costs (2020$) by relapse and payor

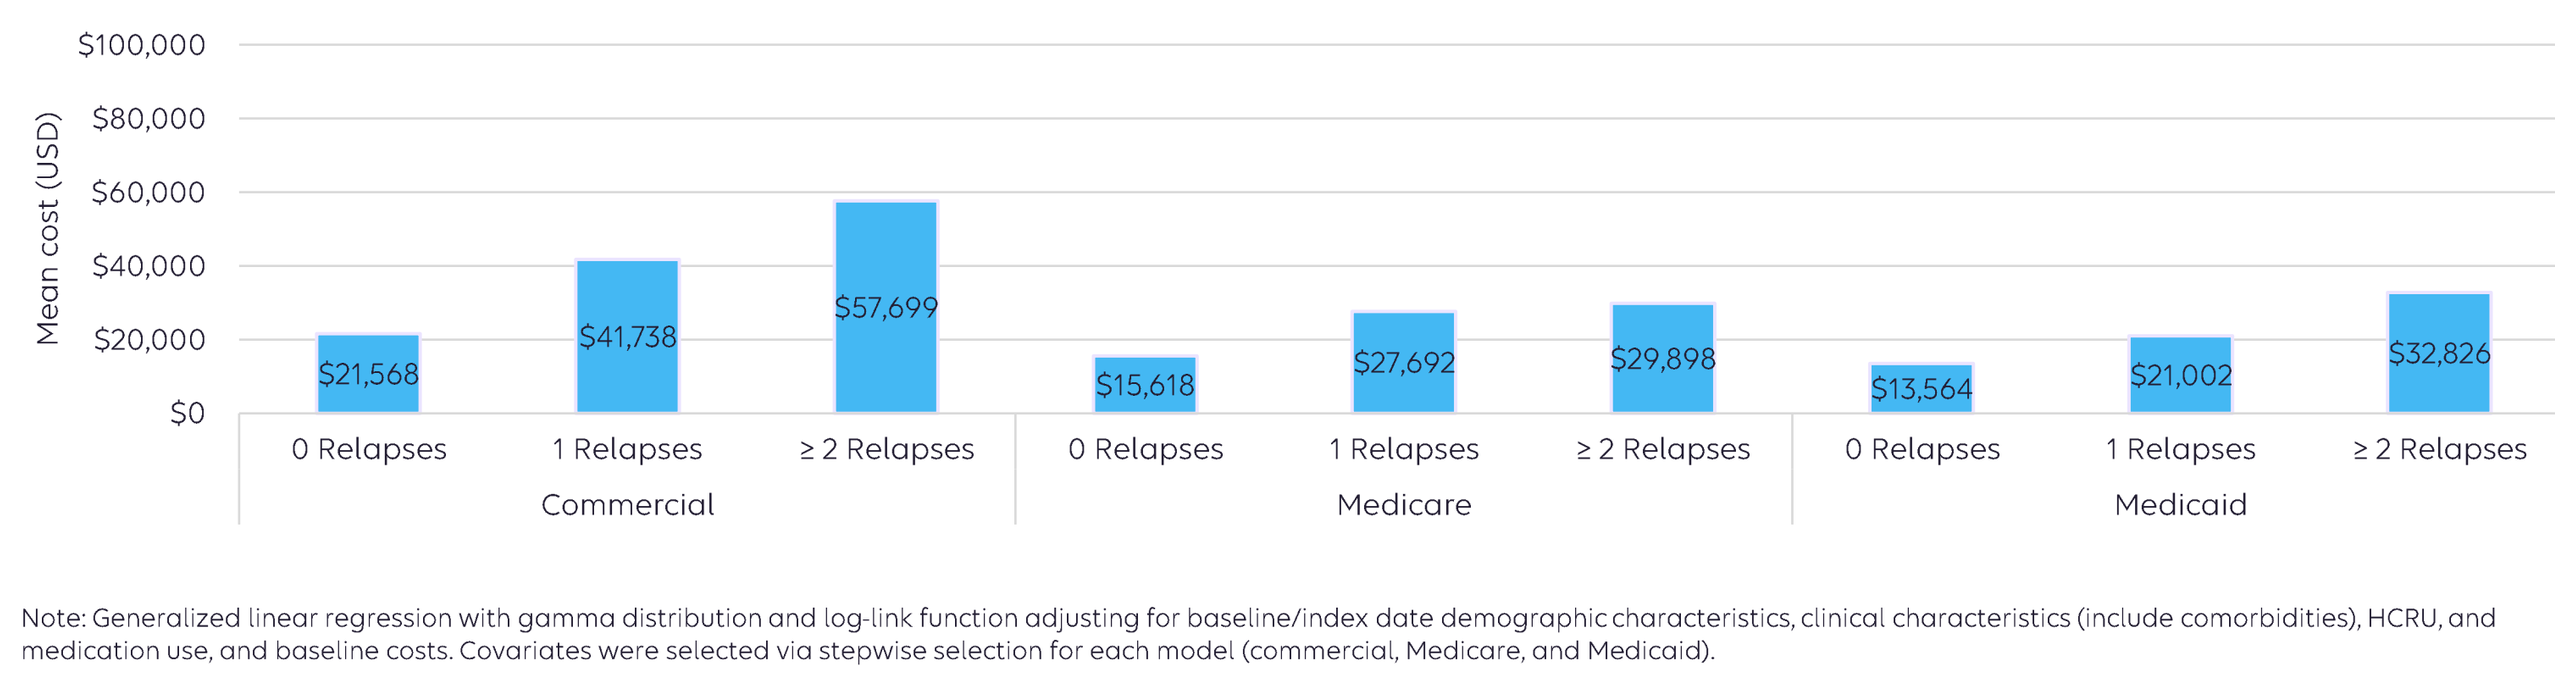
Mean cost (USD)
$100,000
$80,000
$60,000
$40,000
$20,000
$0
$21,568
$41,738
$57,699
$15,618
$27,692
$29,898
$13,564
$21,002
$32,826
0 Relapses
1 Relapses
≥ 2 Relapses
Commercial
Medicare
Medicaid

Note: Generalized linear regression with gamma distribution and log-link function adjusting for baseline/index date demographic characteristics, clinical characteristics (include comorbidities), HCRU, and medication use, and baseline costs. Covariates were selected via stepwise selection for each model (commercial, Medicare, and Medicaid).

Limitations

- Administrative insurance claims are collected for the purpose of payment and not for research and may not reflect a member's full treatment history or true diagnoses as coding issues may occur and medications may not be taken as prescribed.
- Although we attempted to identify patients with incident disease, because of the limited patient history available in the claims, there is the possibility that the first recorded claim in the HIRD was not truly the onset date of the disease.
- Relapse events may be under captured if patients were not seen in emergency room or inpatient settings.
- The cohorts defined by the number of relapses (0 vs. 1 vs. ≥2 relapses) were informed by prior research and clinical guidance; however, there is no consensus in the literature as to what categories to use for frequent relapse.

Conclusions

- Patients with schizophrenia or schizoaffective disorder have high HCRU and costs, which increase with relapse frequency.
 - HCRU and costs nearly doubled between 0 relapse and 2 relapse cohorts in both baseline and post-index periods.
 - Costs were highest in commercial patients and lowest among Medicaid patients, despite similar utilization, likely driven by differences in reimbursement rates.
- Psychotherapy use varied by payor and number of relapses but was suboptimal for the majority of patients.
- This illustrates the high burden and unmet need for managing this disease with opportunities to improve care for these underserved patients.

References

1. Wallace A, Barron J, York W, et al. *J Manag Care Spec Pharm*. 2019;25(10):1102-1110.
2. Wallace A, Isenberg K, York W, et al. *Schizophr Res*. 2020;215:392-398.
3. Huxley P, Krayer A, Poole R, Prendergast L, Aryal S, Warner R. *Brain Behav*. 2021;11(6):e02172.
4. Waters HC, Ruetsch C, Tkacz J. *Am J Manag Care*. 2019;25(12):e373-e378.

Disclosure

LNP, YY, CT, CLC, and JLS were employees of Carelon Research at the time of the study, which received funding from Boehringer Ingelheim Pharmaceuticals, Inc., for the conduct of the study on which this abstract is based. JLS is also an Elevance Health shareholder. TG and PX are employees of Boehringer Ingelheim Pharmaceuticals, Inc. KI is an Elevance Health employee and shareholder.

Poster presented at ISPOR, May 7-10, 2023, Boston, MA, USA.

Pin Xiang | pin.xiang@boehringer-ingelheim.com